Maternal Outcomes in Pregnant Women Receiving Live Attenuated Influenza Vaccine

Seth L. Toback, MD;[1] Richard Beigi, MD;[2] Patricia Tennis, PhD;[3] Frangiscos Sifakis, PhD, MPH;[1] Brian Calingaert, MS;[3] Christopher S. Ambrose, MD[1]

[1]MedImmune, LLC, Gaithersburg, MD, USA; [2]Magee-Womens Hospital, Pittsburgh, PA, USA; [3]RTI Health Solutions, Research Triangle Park, NC, USA

Background

- Because of the increased risk of influenza complications among pregnant women, influenza vaccination during pregnancy is recommended by the Advisory Committee on Immunization Practices.[1]
- As with all live vaccines, live attenuated influenza vaccine (LAIV) is not recommended for use during pregnancy.[1,2]
- Although LAIV is not recommended for use in pregnant women, inadvertent administration of LAIV to pregnant women does occur.
- There are limited data regarding maternal outcomes after LAIV administration during pregnancy.
 - A 2010 analysis of data from the Vaccine Adverse Events Reporting System (VAERS) reported 27 pregnant women from 2003 through 2009 who received LAIV.
 - 60% of the pregnant women did not report any associated adverse event.
 - Of those reporting adverse events in VAERS, the most common were spontaneous abortion (n=3), fever (n=3), vomiting (n=3), headache (n=3), and sore throat (n=2).[3]
- LAIV is approved in the United States for use in eligible children and adults 2 to 49 years of age.
- More than 39 million doses of seasonal trivalent LAIV have been distributed for use in the United States from licensure in 2003 through February 2011.[4]

Objectives

- To estimate the rate of LAIV use among pregnant women
- To describe maternal outcomes after vaccination with LAIV

Methods

- Anonymized, person-level data from The LifeLink™ Health Plan Claims Database (IMS Health, Norwalk, CT) that covers approximately 50 million individuals were analyzed for the 6 influenza seasons from 2003–2004 through 2008–2009 to assess maternal outcomes after LAIV vaccination.
- Inclusion criteria included female gender, age (12–49 y), claim for the delivery of a child, and having at least 270 days of continuous enrollment in the database before delivery.
- Cohort characteristics were analyzed using descriptive statistics.
- The proportion of deliveries with LAIV vaccination during pregnancy was calculated by dividing the number of eligible deliveries from women who had been vaccinated with LAIV by the total number of eligible deliveries that occurred on or after October 1, 2003, through September 2009.
- Because the delivery of a child was an entry criterion and delivery data were only available through September 2009, the 2008–2009 cohort would not include first trimester vaccinations of women with deliveries after September 2009.
- Emergency department (ED) visits and hospitalizations occurring within 42 days of vaccination were analyzed by primary diagnosis; outcomes were categorized as cardiopulmonary, obstetric, and other.
- The primary discharge diagnoses associated with any hospitalization from the time of vaccination up until, but not including, the hospitalization at the time of delivery were collected and analyzed.
- The database was unable to link maternal claims data to the claims data describing their offspring; thus, no information regarding birth outcomes was available.

Results

- A total of 834,999 women were identified with a pregnancy resulting in a delivery between October 2003 and September 2009.
 - 138 women (0.017%) had a claim for vaccination with LAIV while pregnant.
- The number of vaccinations during pregnancy increased throughout the study period **(Table 1)**.

Table 1. Number of Vaccinations With LAIV During Pregnancy: 2003–2004 to 2008–2009 Influenza Seasons

Season	Vaccinations During Pregnancy, n
2003–2004	14
2004–2005	12
2005–2006	17
2006–2007	32
2007–2008	22
2008–2009	41

LAIV=live attenuated influenza vaccine.

- Since the 2004–2005 season, the rate of LAIV vaccination during pregnancy has remained relatively constant with a range of 5.7–12.3 per million LAIV doses distributed per season.
- The characteristics of those vaccinated with LAIV versus those not vaccinated with LAIV were generally comparable **(Table 2)**.
- Of those vaccinated with LAIV, 47%, 27%, and 26% were vaccinated in the first, second, and third trimesters, respectively.
 - Among the 65 first trimester exposures, 42 occurred during the first 6 weeks of pregnancy.
- When trimester of vaccination was analyzed by influenza season, no trends were discernible among the small number of vaccinations occurring each season **(Figure 1)**.

Figure 1. Distribution of the Trimester of Vaccination With LAIV

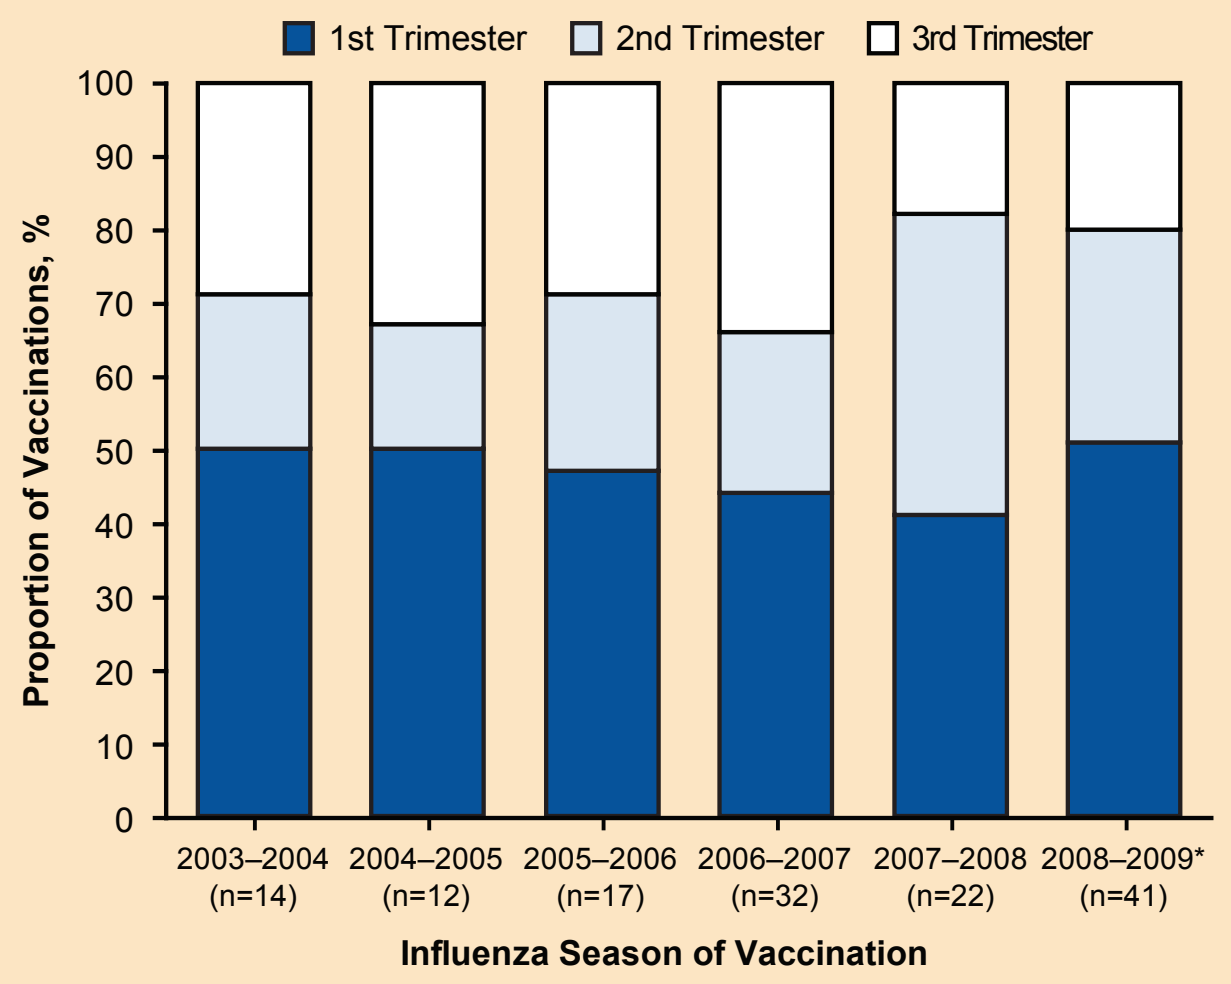
1st Trimester
2nd Trimester
3rd Trimester
Proportion of Vaccinations, %
100
90
80
70
60
50
40
30
20
10
0
2003–2004 (n=14)
2004–2005 (n=12)
2005–2006 (n=17)
2006–2007 (n=32)
2007–2008 (n=22)
2008–2009* (n=41)
Influenza Season of Vaccination

LAIV=live attenuated influenza vaccine.
*Because the data analyzed included deliveries through September 2009, the 2008–2009 cohort may be missing some first trimester vaccinations.

- Eight unique individuals were found to have a claim for a hospitalization or ED visit within 42 days of vaccination with LAIV (3 hospitalizations, 5 ED visits; **Table 3**).
 - Overall, 5.8% of women experienced a hospitalization or ED visit within 42 days after vaccination with LAIV.
 - 1.4%, 0.7%, and 3.6% experienced obstetric, cardiopulmonary, and other medical events, respectively **(Table 3)**.
- Five additional hospitalizations that occurred more than 42 days postvaccination were noted before delivery **(Table 4)**.

Table 2. Characteristics of the Vaccinated With LAIV and Not Vaccinated With LAIV Cohorts

	Vaccinated With LAIV (n=138) n (%)	Not Vaccinated With LAIV (n=834,861) n (%)
Maternal age at delivery, y		
14–19	18 (13)†	45,447 (5)
20–34	93 (67)	629,545 (75)
35–43	27 (20)	159,869 (19)
Gestational age at birth, wk		
≤36	7 (5)	66,003 (8)
>36	131 (95)	768,858 (92)
High-risk medical condition*		
Cardiac disease	5 (4)	19,172 (2)
Pulmonary disease	10 (7)	42,491 (5)
Diabetes mellitus	7 (5)	25,649 (3)
Renal disease	0 (0)	1096 (0)
Malignancy	1 (1)	4752 (1)
Immunosuppresive disorder	0 (0)	444 (0)
Any high-risk condition	21 (15)	86,666 (10)
Claim for TIV during pregnancy	6 (4)†	96,600 (12)
Prenatal visit claims during pregnancy		
0	7 (5)	48,041 (6)
1–3	33 (24)	225,294 (27)
4–6	43 (31)	291,776 (35)
7–10	26 (19)	164,468 (20)
>10	29 (21)‡	105,282 (13)
Trimester at vaccination		
1st	65 (47)	NA
2nd	37 (27)	NA
3rd	36 (26)	NA

LAIV=live attenuated influenza vaccine; NA=not applicable; TIV=trivalent inactivated influenza vaccine.
*At least 1 hospital/ED claim or 2 outpatient claims any time on or before delivery date.
†*P*<0.0001 vs not vaccinated with LAIV cohort.
‡*P*<0.01 vs not vaccinated with LAIV cohort.

Table 3. Primary Diagnoses for Emergency Department Visit or Hospitalization Within 42 Days After LAIV Vaccination

Event	Events* Among Women Vaccinated With LAIV, n: ED Visit	Hosp	ED Visit or Hosp	Proportion of Women Vaccinated With LAIV with ED Visit or Hosp, %
Any complication	5	3	8	5.8
Obstetrical complications				
Any	0	2	2	1.4
643.13 Hyperemesis gravidarum with metabolic disturbance, antepartum	0	1	1	0.7
644.03 Threatened premature labor, without delivery	0	1	1	0.7
Cardiopulmonary conditions				
Any	1	0	1	0.7
466.0 Acute bronchitis	1	0	1	0.7
Other conditions				
Any	4	1	5	3.6
590.80 Pyelonephritis, unspecified	0	1	1	0.7
789.06 Epigastric symptoms involving abdomen and pelvis	1	0	1	0.7
729.5 Pain in limb	1	0	1	0.7
787.91 Diarrhea	1	0	1	0.7
786.59 Chest pain; other	1	0	1	0.7

ED=emergency department; Hosp=Hospitalization; LAIV=live attenuated influenza vaccine.
*Excludes visits for delivery. Each woman could contribute only 1 ED visit and 1 hospitalization per ICD-9 code.

Table 4. Listing of Primary Diagnoses Associated With All Hospitalizations Occurring Any Time Between LAIV Vaccination and Delivery*

Diagnosis	Day of Occurrence†	Length of Stay, d
Within 42 days postvaccination		
590.80 Pyelonephritis, unspecified	6	3
643.13 Hyperemesis gravidarum with metabolic disturbance, antepartum	7	3
644.03 Threatened premature labor, without delivery	9, 12‡	1
More than 42 days postvaccination		
233.1 Carcinoma in situ of cervix uteri	51	1
590.1 Acute pyelonephritis	62§	5
656.73 Other placental conditions affecting management of mother, antepartum	126	4
652.23 Breech presentation without version, antepartum	184	1
487.1 Influenza with other respiratory manifestations	240	1

LAIV=live attenuated influenza vaccine.
*Excludes hospitalizations for delivery.
†Day of occurrence relative to LAIV vaccination (1=day of vaccination).
‡Both hospitalizations for code 644.03 occurred in the same subject on separate days.
§Same subject hospitalized for diagnosis code 590.80 on day 6 of occurrence.

Conclusions

- **Administration of LAIV to pregnant women is rare and the rate per million doses distributed has remained relatively constant since 2004–2005.**
- **LAIV use in pregnancy is most likely to occur very early in the pregnancy, when a pregnancy may be unrecognized. However, use later in pregnancy was also observed, suggesting that some healthcare providers would benefit from increased education regarding the recommended use of LAIV.**
- **In this cohort, there was no evidence of significant maternal adverse outcomes after receipt of LAIV.**
- **All outcomes identified after LAIV exposure occurred at rates similar to rates in unvaccinated pregnant women reported in the medical literature.[5-11]**
- **These data provide safety information to providers and pregnant women in the event of inadvertent LAIV administration, but do not support the routine use of LAIV in pregnant women.**

References

1. Fiore AE, et al. MMWR Recomm Rep. 2010;59:1-62.
2. Centers for Disease Control and Prevention. Guidelines for Vaccinating Pregnant Women. Available at: http://www.cdc.gov/vaccines/pubs/preg-guide.htm. Accessed February 22, 2011.
3. Moro PL, et al. Am J Obstet Gynecol. 2010;204:146.e1-7.
4. Data on File. MedImmune, LLC, Gaithersburg, MD.
5. Banhidy F, et al. Eur J Epidemiol. 2008;23:29-35.
6. Bennett TA, et al. Am J Obstet Gynecol. 1998;178:346-354.
7. Jolley JA and Wing DA. Drugs. 2010;70:1643-1655.
8. McPheeters ML, et al. Am J Obstet Gynecol. 2005;192:1325-1329.
9. Millar LK and Cox SM. Infect Dis Clin North Am. 1997;11:13-26.
10. Nichol KL, et al. JAMA. 1999;282:137-144.
11. Verberg MF, et al. Hum Reprod Update. 2005;11:527-539.

Sponsored by MedImmune, LLC.

45th National Immunization Conference • March 28–30, 2011 • Washington DC, USA